110

Comparison of cost per response between deucravacitinib and apremilast/biologics in patients with moderate to severe plaque psoriasis

Sang Hee Park,[1] Mark Lambton,[2] Jordana Schmier,[3] Dipen Patel,[3] Sara Hovland,[1] Keith Wittstock,[1] Vardhaman Patel[1]

[1]Bristol Myers Squibb, Princeton, NJ, USA; [2]OPEN Health Evidence & Access, York, UK; [3]OPEN Health Evidence & Access, Parsippany, NJ, USA

Background

- The average cost of treatment for psoriasis (PsO) in US patients is high, with per-patient per-year costs estimated to be $12,523[1]
 - Healthcare costs are estimated to be 2.5 times higher for patients with moderate to severe PsO than those for patients with mild PsO[1]
- Deucravacitinib, an oral, selective, allosteric tyrosine kinase 2 (TYK2) inhibitor, is approved by the US Food and Drug Administration for the treatment of adults with moderate to severe plaque PsO who are candidates for systemic therapy or phototherapy[2]
- In the pivotal clinical trials POETYK PSO-1 and PSO-2, greater proportions of patients treated with deucravacitinib achieved static Physician's Global Assessment scores of 0 or 1 (0/1) and/or 75% reduction in baseline Psoriasis Area and Severity Index score (PASI 75) than those treated with placebo or apremilast at Week 16[2]

Objectives

Primary

- To estimate and compare the cost per response (CPR) of deucravacitinib vs apremilast for patients with moderate to severe PsO from a US commercial payer perspective

Secondary

- To estimate and compare the CPR of deucravacitinib vs first-line (1L) branded systemic treatments for patients with moderate to severe PsO from a US commerial payer perspective

Methods

- A model was developed in Microsoft Excel to assess CPR from a US commercial payer perspective (**Figure 1**)
- CPR was compared between deucravacitinib and apremilast/1L branded systemics across 2 time frames:
 - **Short-term (16 or 24 weeks)**, assuming patients continue treatment for 16 or 24 weeks after initiation with deucravacitinib or apremilast/1L branded systemics
 - **Long-term (52 weeks)**, assuming patients continue index treatment if they achieve response at 16 or 24 weeks, or switch from index to second-line (2L) biologic therapy if they do not achieve response at 16 or 24 weeks
- The number needed to treat (NNT), representing the number of patients who would need to be treated with a less efficacious treatment vs a more efficacious treatment to achieve one additional patient response, was estimated as an exploratory endpoint

Figure 1. Model overview (long-term)

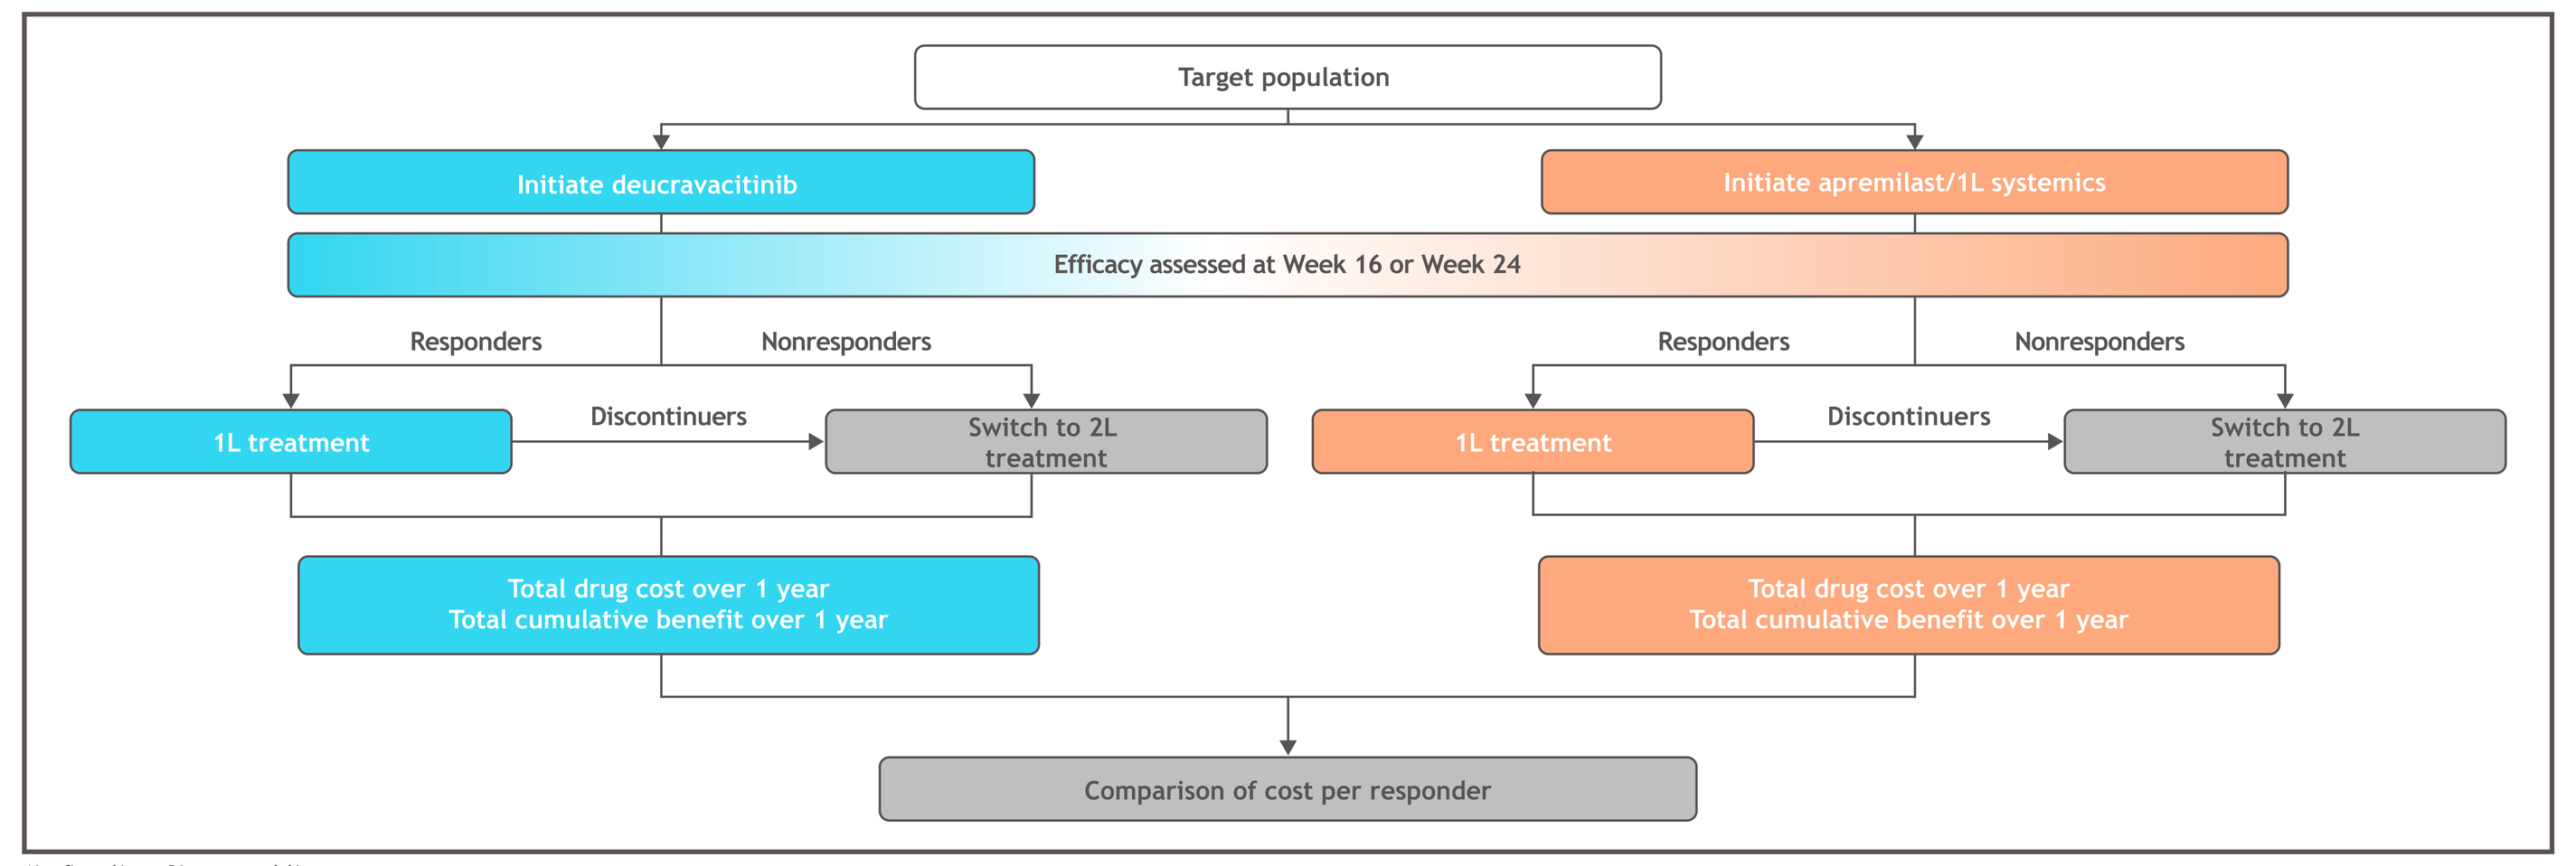

Target population
Initiate deucravacitinib
Initiate apremilast/1L systemics
Efficacy assessed at Week 16 or Week 24
Responders
Nonresponders
1L treatment
Discontinuers
Switch to 2L treatment
Total drug cost over 1 year
Total cumulative benefit over 1 year
Comparison of cost per responder

1L, first-line; 2L, second-line.

Model assumptions

- Pharmacy (wholesale acquisition costs, derived from Merative™ MicroMedex® Red Book, September 2022) and administration costs (based on Practice Management Information Costs Medical Fees 2021) are the only costs accounted for in the model
- All patients receive treatment according to prescribing information and adhere 100% to assigned treatment

Definition of response

- **Short-term:** achieving sPGA 0/1 or PASI 75 at 16 or 24 weeks for deucravacitinib vs apremilast, and PASI 75 at 16 or 24 weeks for deucravacitinib vs 1L branded systemics
 - Deucravacitinib vs apremilast: based on pooled efficacy demonstrated by deucravacitinib and apremilast in POETYK PSO-1 and PSO-2[2]
 - Deucravacitinib vs 1L branded systemics: based on a weighted average of treatment efficacy shown in a network meta-analysis (NMA)[3] and respective market share
 - Market share for 1L branded systemics was based on real-world market share distribution estimated in an internal claims analysis (**Figure 2**)
- **Long-term:** cumulative time spent in PASI 75 response over 52 weeks,[4,5] with efficacy assessed at Week 16 or Week 24; nonresponders were assumed to switch to subsequent treatment based on market share (**Figure 3**)
 - Measured as the area under the curve of PASI 75 response over 52 weeks using the trapezoidal rule
 - Assumes a linear increase in percentage of patients responding
 - Deucravacitinib vs apremilast: based on pooled efficacy demonstrated by deucravacitinib and apremilast in POETYK PSO-1 and PSO-2[2,6]
 - Deucravacitinib vs 1L branded systemics: based on an NMA of treatment efficacy[3]

Treatment discontinuation

- For the long-term analysis of deucravacitinib vs apremilast, in addition to treatment switch in the event of failure to respond at 16 or 24 weeks, index treatments were assumed to incur a discontinuation rate after Week 16 or 24, based on the rates observed in the POETYK PSO-1 and PSO-2 trials (4.4% for deucravacitinib; 11.6% for apremilast)
- For the long-term analysis of deucravacitinib vs 1L branded systemics, in addition to treatment switch in the event of failure to respond at 16 or 24 weeks, index treatments were assumed to incur a discontinuation rate after Week 16 or 24, based on real-world discontinuation rates[7]
 - The discontinuation rate for 1L branded systemics was 22.4%
 - As there is no current real-world discontinuation rate for deucravacitinib, branded oral treatments were assumed to have the same discontinuation rate (16.5%)
- Patients who discontinued were assumed to switch to 2L biologic treatment

Figure 2. US market share distribution for 1L branded systemics for PsO[a]

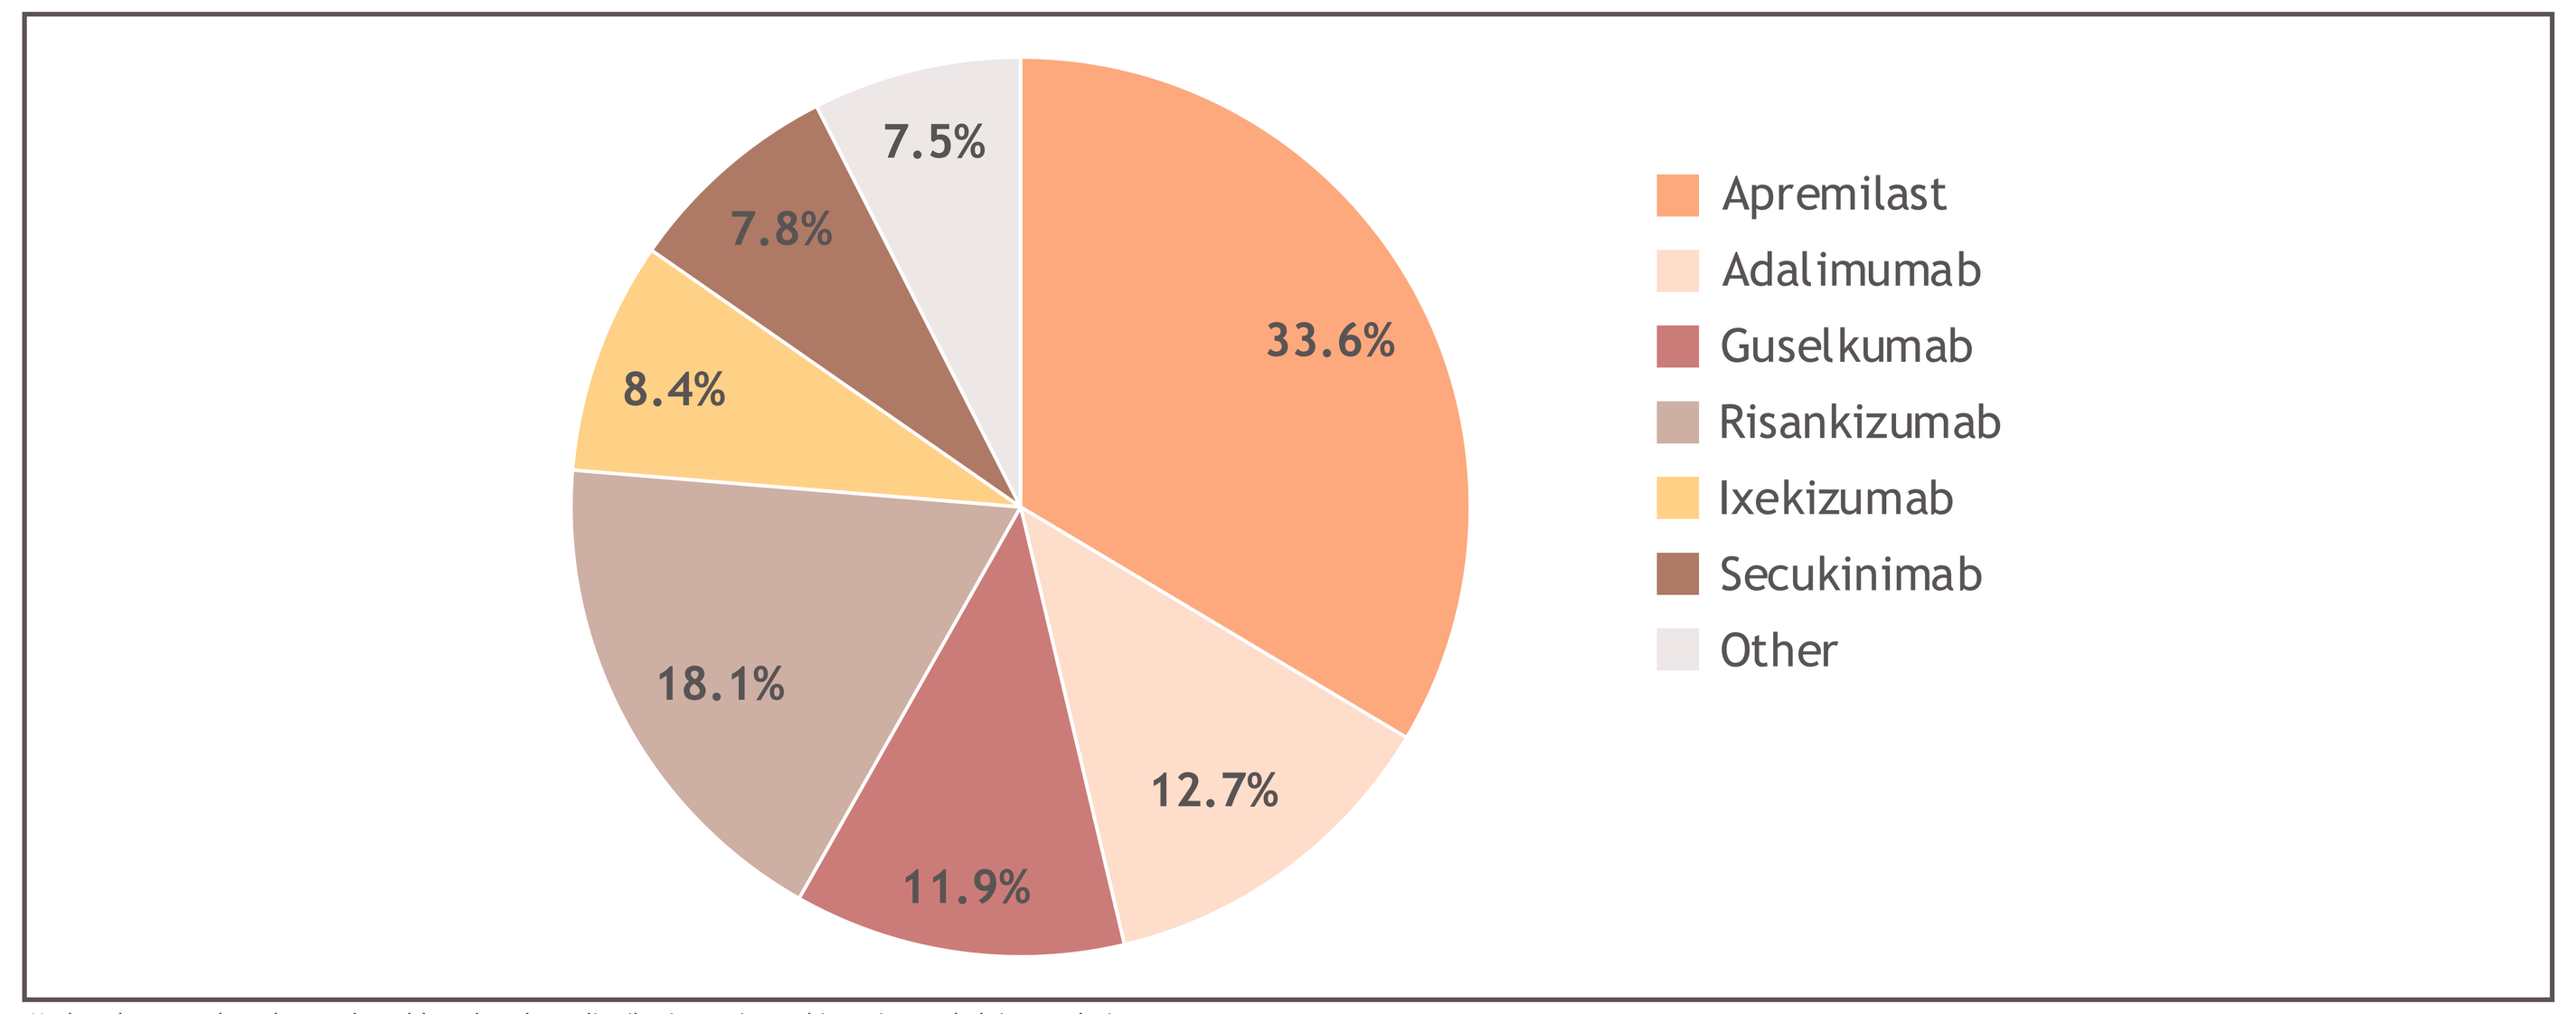

33.6%
12.7%
11.9%
18.1%
8.4%
7.8%
7.5%
Apremilast
Adalimumab
Guselkumab
Risankizumab
Ixekizumab
Secukinimab
Other

[a]Market share was based on real-world market share distribution estimated in an internal claims analysis.
1L, first-line; PsO, psoriasis.

Figure 3. US market share distribution for 2L biologic treatments[a]

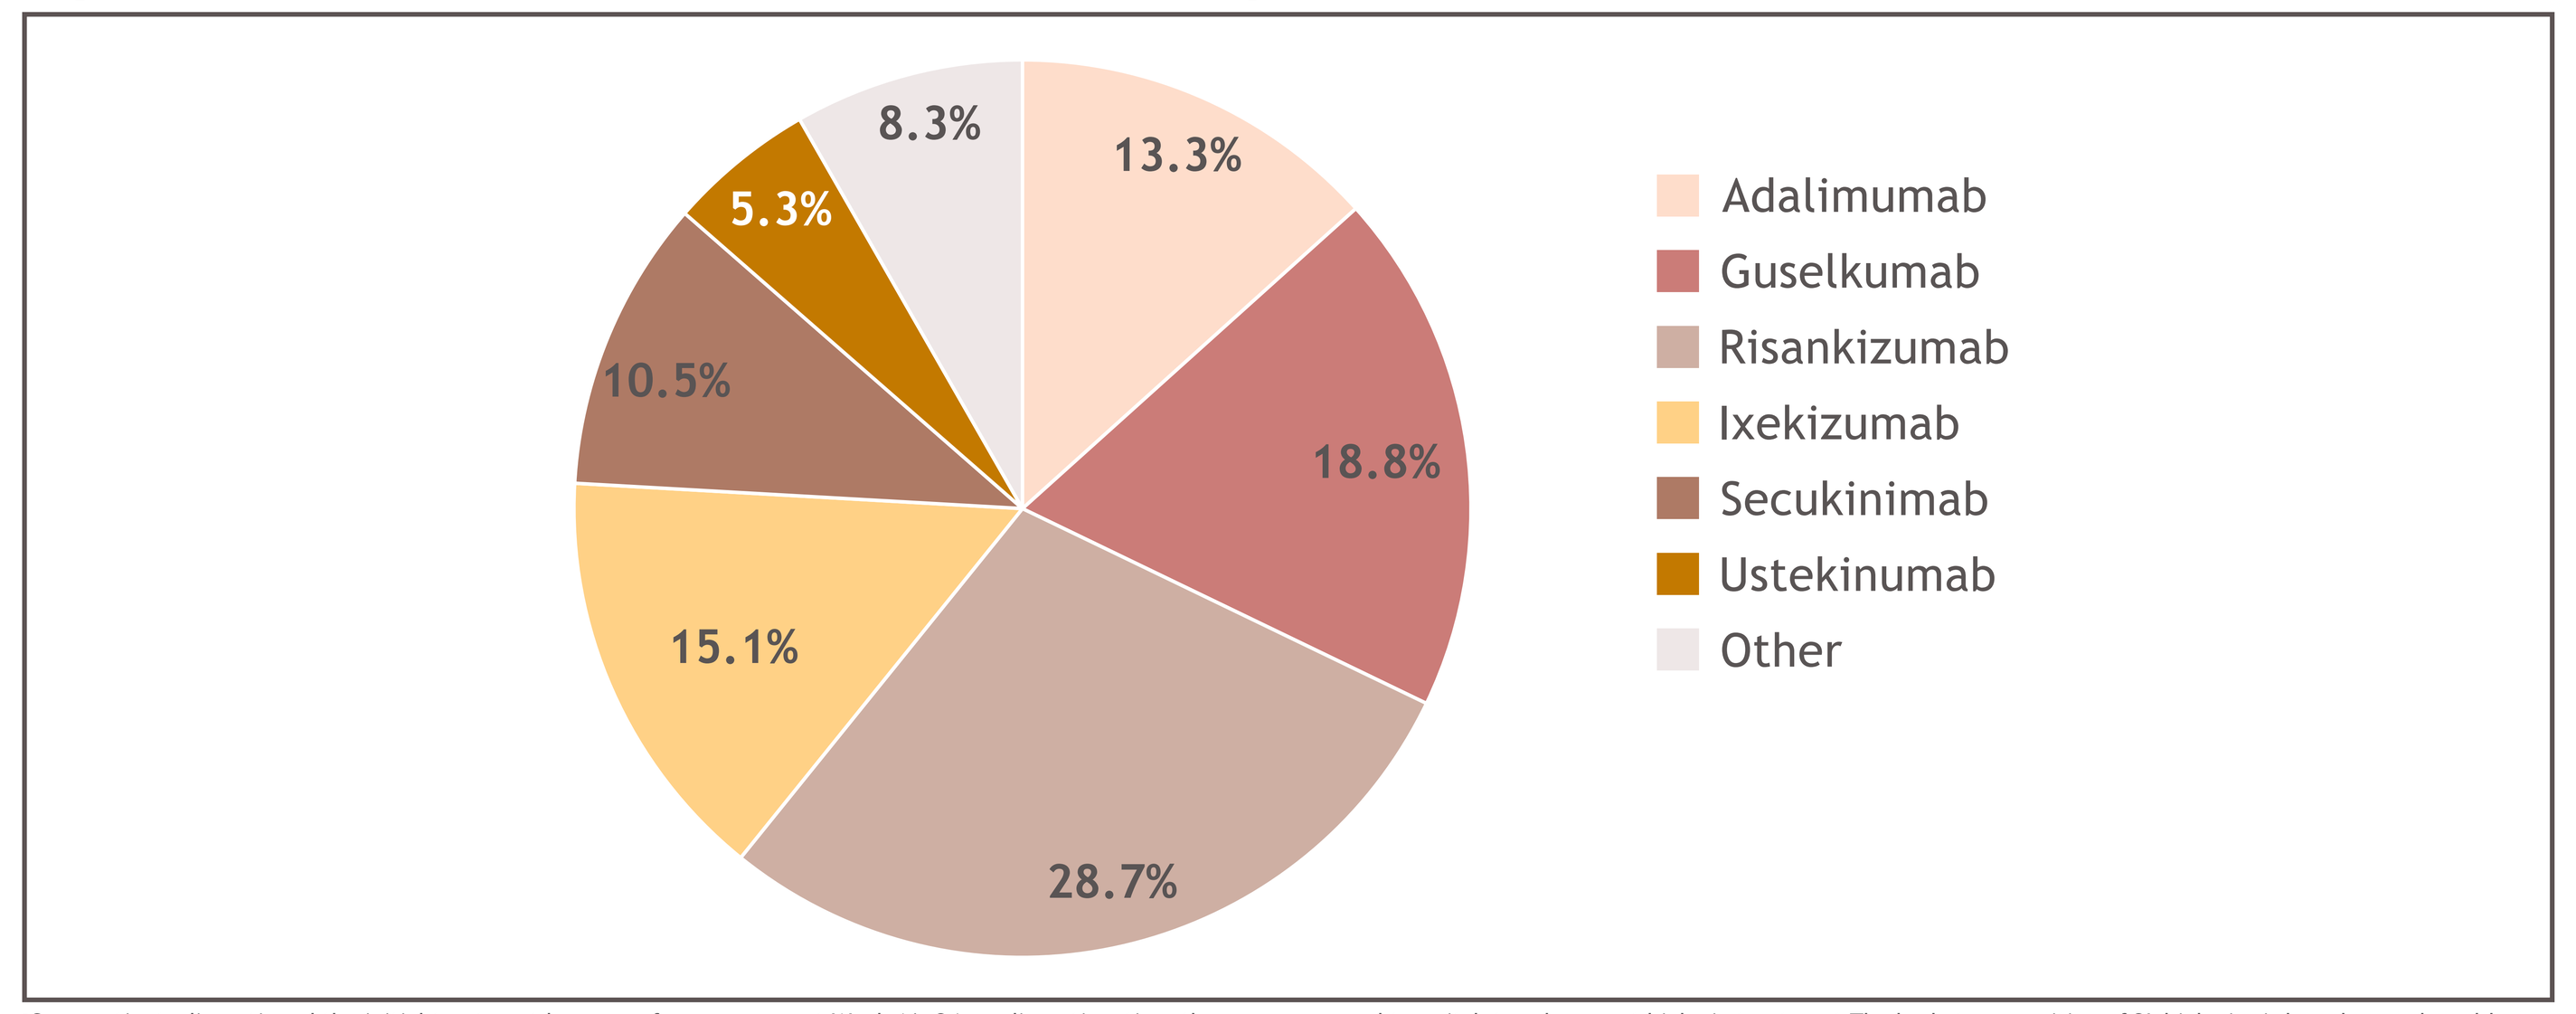

13.3%
18.8%
28.7%
15.1%
10.5%
5.3%
8.3%
Adalimumab
Guselkumab
Risankizumab
Ixekizumab
Secukinimab
Ustekinumab
Other

[a]Once patients discontinued the initial treatment because of nonresponse at Week 16, 24, or discontinuation, they were assumed to switch to subsequent biologic treatment The basket composition of 2L biologics is based on real-world market share distribution, estimated in an internal claims analysis
2L, second-line.

Definitions and formulas

- **CPR for short-term:** Calculated with total cost and clinical response (defined as achieving sPGA 0/1 or PASI 75 at 16 or 24 weeks for deucravacitinib vs apremilast, and PASI 75 for deucravacitinib vs 1L branded systemics)

$$\frac{\text{Cost per patient at the end of time horizon}}{\text{Clinical response rate at the end of time horizon}}$$

- **CPR for long-term:** Calculated with total cost and cumulative PASI 75 response over 52 weeks

$$\frac{\text{Cost per patient over 52 weeks}}{\text{Cumulative clinical response over 52 weeks}}$$

- **Difference in CPR** = CPR of deucravacitinib − CPR of comparators
- **NNT** = $\frac{1}{\text{Treatment 1 (higher response rate)} - \text{Treatment 2 (lower response rate)}}$

Scenario analyses

- Comparator treatment prices increase by 10% (deucravacitinib price remains the same)
- Discontinuation rate set to 0 for all treatments
- Cumulative response over 52 weeks with all patients receiving only one 2L treatment: (a) adalimumab, (b) risankizumab, (c) guselkumab, (d) ixekizumab, or (e) secukinumab

Results

Deucravacitinib vs apremilast

- At Week 16, patients initiating deucravacitinib had a numerically or statistically significantly lower CPR than apremilast initiators (**Figure 4**)
 - sPGA 0/1: −$4347 (95% confidence interval [CI], −$7652 to −$1785)
 - PASI 75: −$1376 (95% CI, −$3734 to $494)
- At Week 24, the difference in CPR was greater than at Week 16, with statistically significant results
 - sPGA 0/1: −$16,039 (95% CI, −$22,364 to −$11,466)
 - PASI 75: −$9099 (95% CI, −$12,997 to −$6090)
- Over 52 weeks, with efficacy assessed at Week 16, CPR was −$3197 lower for deucravacitinib vs apremilast (95% CI, −$6341 to −$492)
- Over 52 weeks, with efficacy assessed at Week 24, the difference in CPR with deucravacitinib was greater, at −$16,090 (95% CI, −$20,988 to −$12,281)

Deucravacitinib vs 1L branded systemics

- At Week 16, patients initiating deucravacitinib had a lower CPR than initiators with 1L branded systemics (−$11,747 [95% CI, −$12,855 to −$10,126]) (**Figure 5**)
- At Week 24, deucravacitinib had a lower CPR than 1L branded systemics: −$14,805 (95% CI, −$15,680 to −$13,438) (**Figure 5**)
- Over 52 weeks, with efficacy assessed at Week 16, CPR was −$4294 lower for deucravacitinib vs 1L branded systemics (95% CI, −$5998 to −$1918)
- Over 52 weeks, with efficacy assessed at Week 24, CPR was −$13,121 with deucravacitinib vs 1L branded systemics (95% CI, −$14,414 to −$11,033)

Figure 4. CPR savings: deucravacitinib vs apremilast

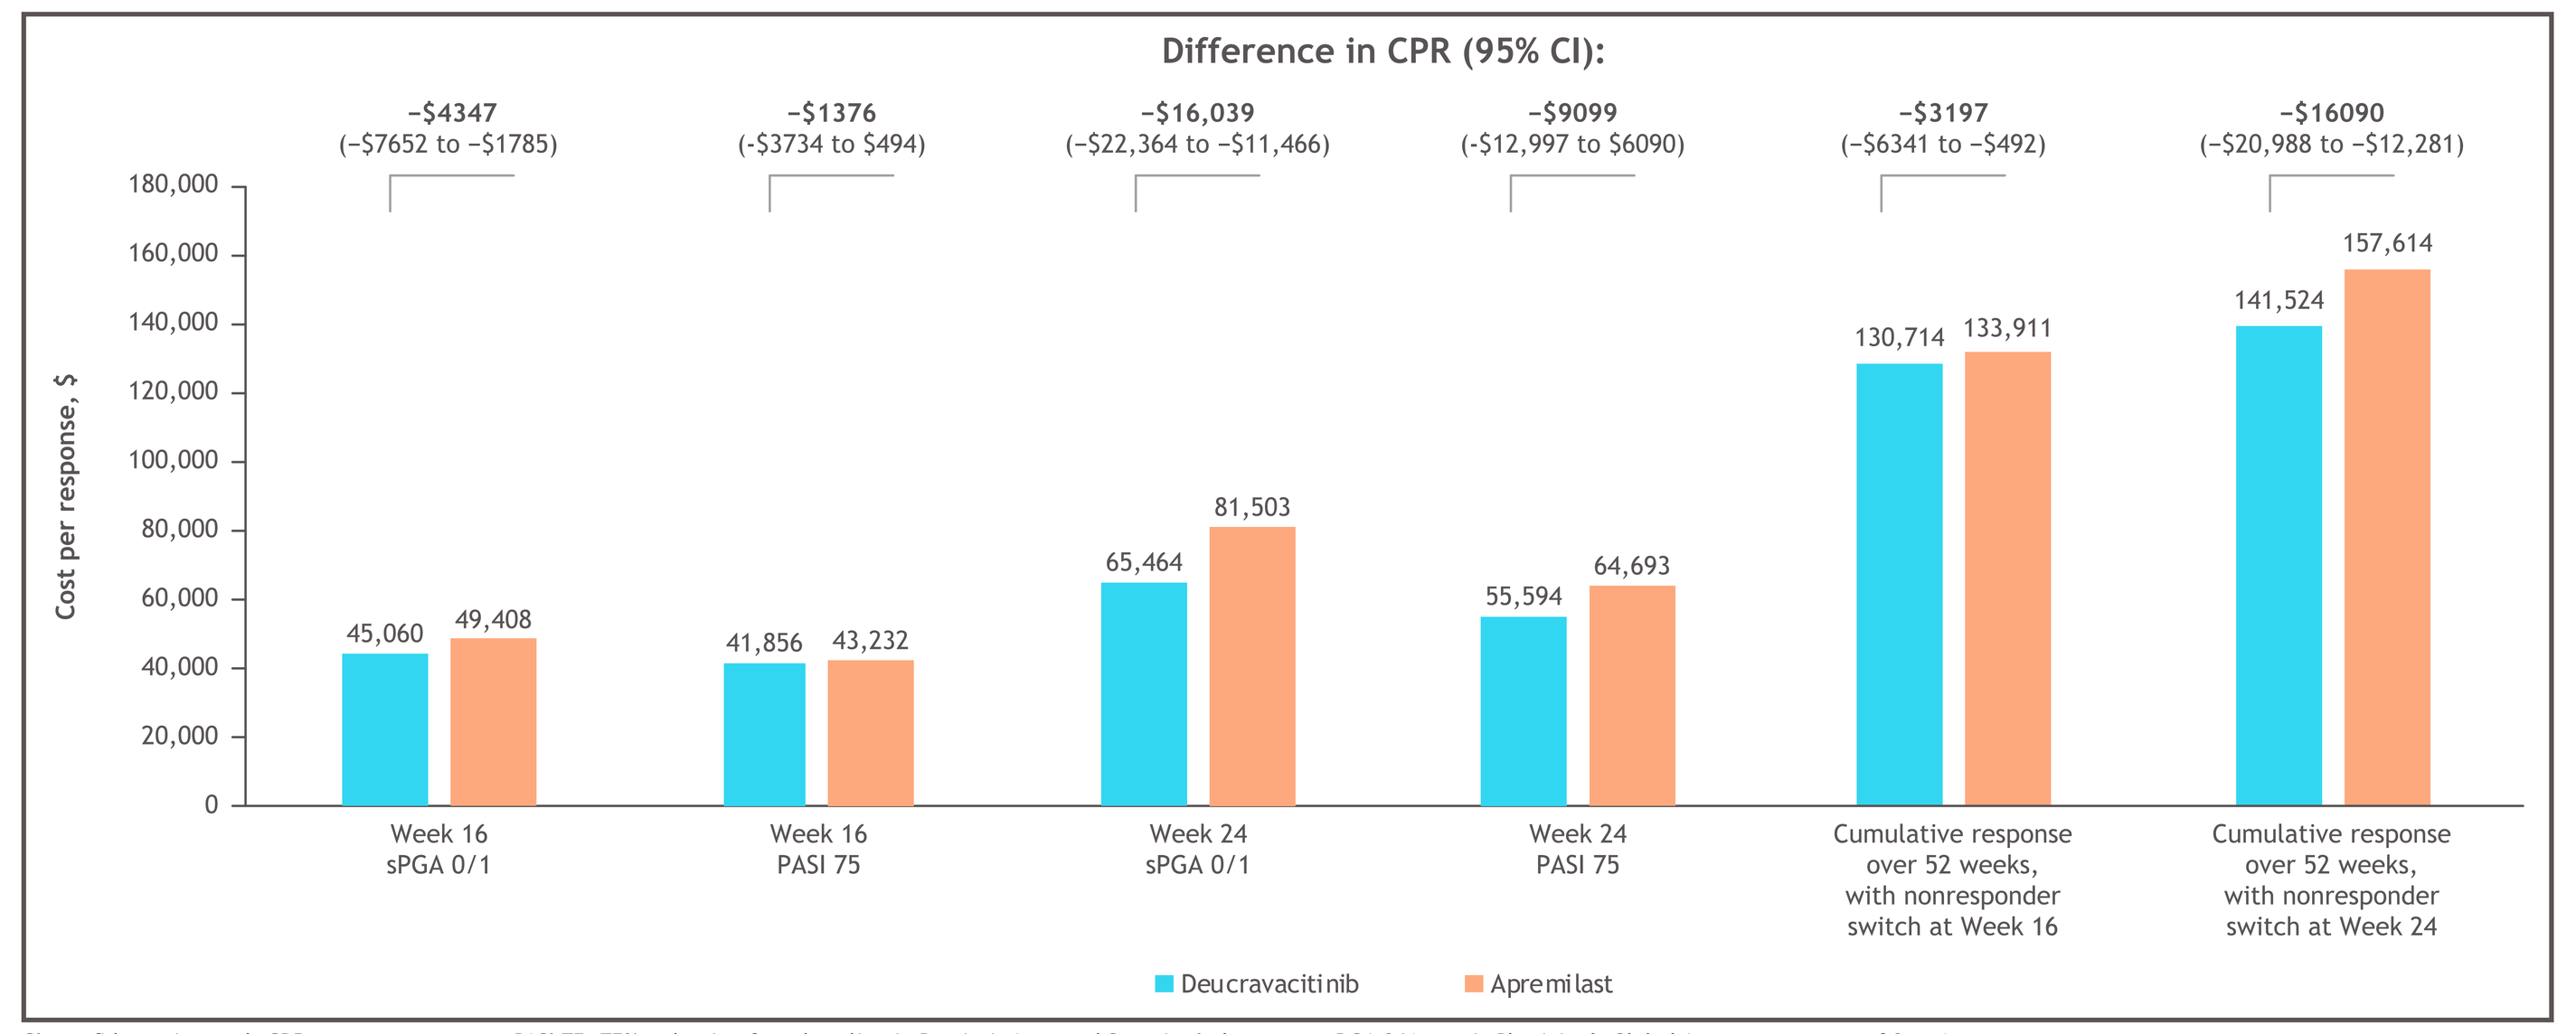

Difference in CPR (95% CI):
−$4347 (−$7652 to −$1785)
−$1376 (−$3734 to $494)
−$16,039 (−$22,364 to −$11,466)
−$9099 (−$12,997 to $6090)
−$3197 (−$6341 to −$492)
−$16090 (−$20,988 to −$12,281)
Cost per response, $
45,060
49,408
41,856
43,232
65,464
81,503
55,594
64,693
130,714
133,911
141,524
157,614
Week 16 sPGA 0/1
Week 16 PASI 75
Week 24 sPGA 0/1
Week 24 PASI 75
Cumulative response over 52 weeks, with nonresponder switch at Week 16
Cumulative response over 52 weeks, with nonresponder switch at Week 24
Deucravacitinib
Apremilast

CI, confidence interval; CPR, cost per response; PASI 75, 75% reduction from baseline in Psoriasis Area and Severity Index score; sPGA 0/1, static Physician's Global Assessment score of 0 or 1.

Figure 5. CPR savings: deucravacitinib vs 1L branded systemics

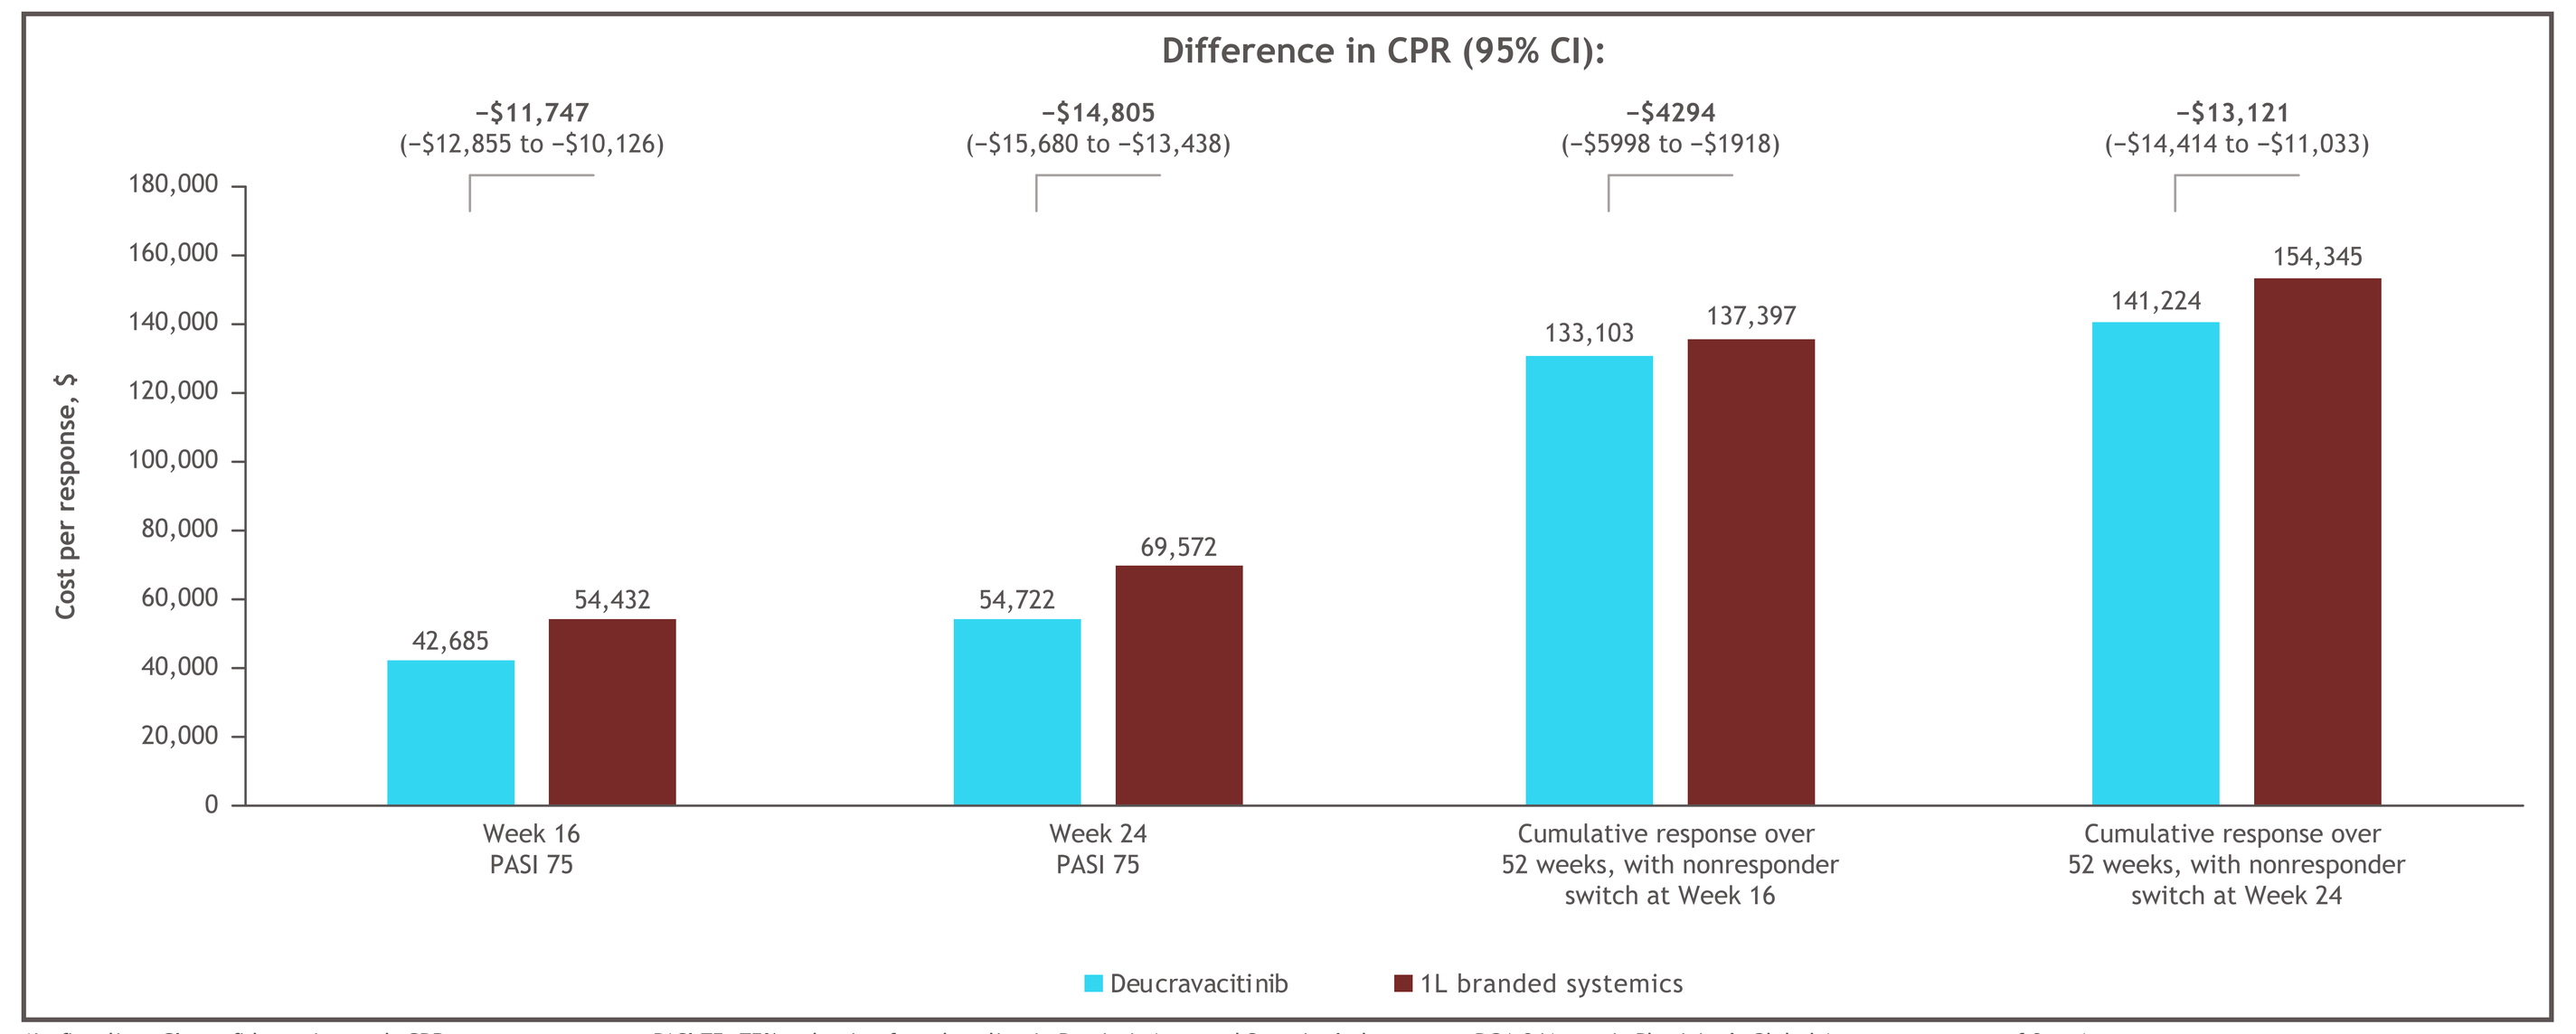

Difference in CPR (95% CI):
−$11,747 (−$12,855 to −$10,126)
−$14,805 (−$15,680 to −$13,438)
−$4294 (−$5998 to −$1918)
−$13,121 (−$14,414 to −$11,033)
Cost per response, $
42,685
54,432
54,722
69,572
133,103
137,397
141,224
154,345
Week 16 PASI 75
Week 24 PASI 75
Cumulative response over 52 weeks, with nonresponder switch at Week 16
Cumulative response over 52 weeks, with nonresponder switch at Week 24
Deucravacitinib
1L branded systemics

1L, first-line; CI, confidence interval; CPR, cost per response; PASI 75, 75% reduction from baseline in Psoriasis Area and Severity Index score; sPGA 0/1, static Physician's Global Assessment score of 0 or 1.

- When compared with apremilast, deucravacitinib had an NNT of 4.1 to 5.8 in the short term (16 or 24 weeks) and 8.8 to 13.7 in the long term (52 weeks) to achieve one additional patient response (**Figure 6**)
- When compared with deucravacitinib, 1L branded systemics had an NNT of 8.5 to 22.7 in the short term, and 22.7 to 55.2 in the long term to achieve one additional patient response (**Figure 7**)

Figure 6. Clinical response for deucravacitinib vs apremilast and NNT for deucravacitinib

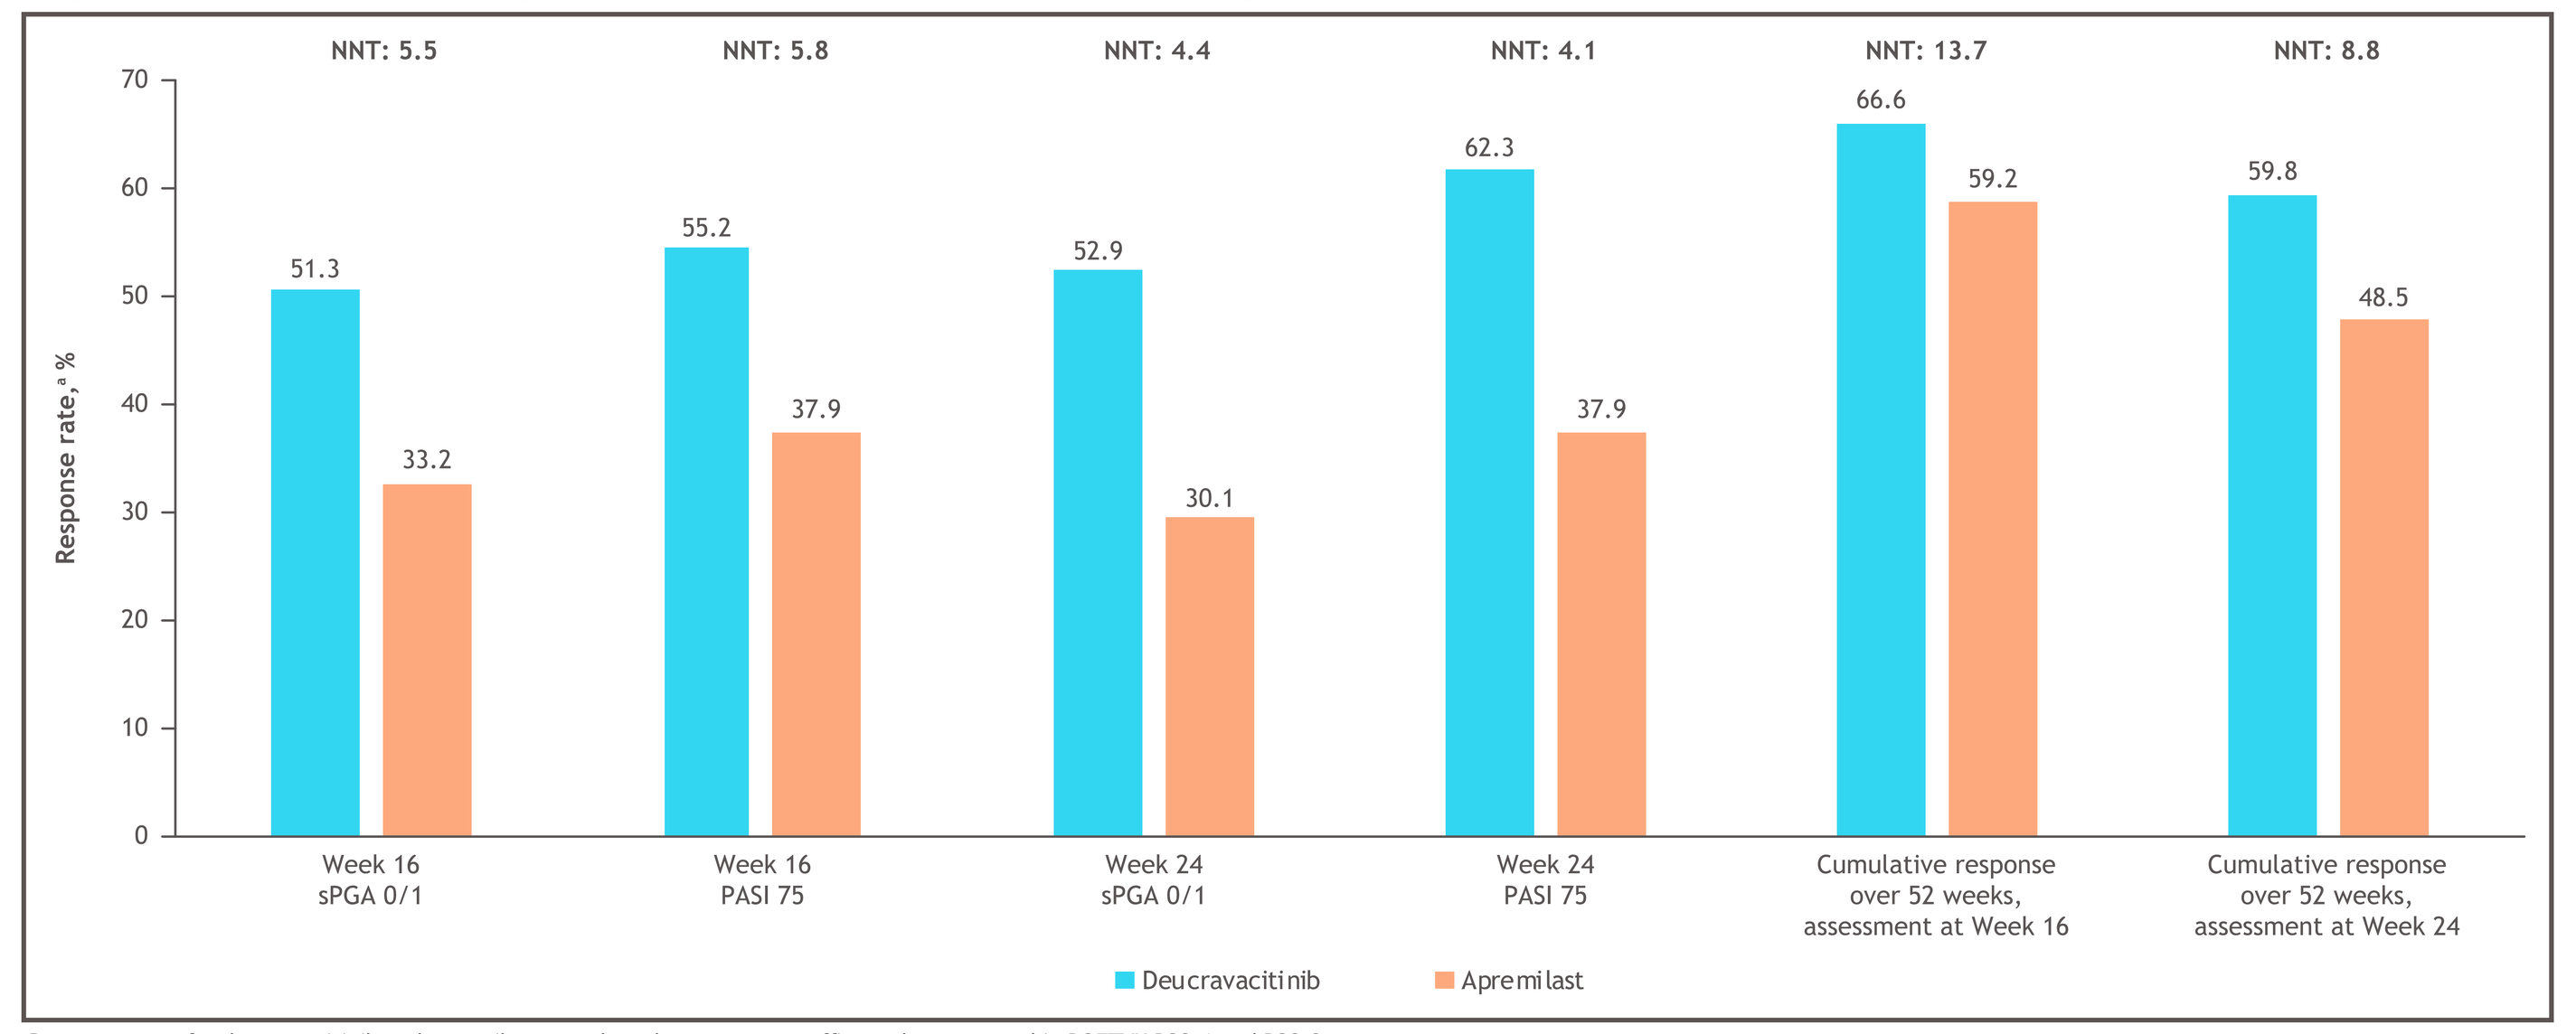

NNT: 5.5
NNT: 5.8
NNT: 4.4
NNT: 4.1
NNT: 13.7
NNT: 8.8
Response rate,[a] %
51.3
33.2
55.2
37.9
52.9
30.1
62.3
37.9
66.6
59.2
59.8
48.5
Week 16 sPGA 0/1
Week 16 PASI 75
Week 24 sPGA 0/1
Week 24 PASI 75
Cumulative response over 52 weeks, assessment at Week 16
Cumulative response over 52 weeks, assessment at Week 24
Deucravacitinib
Apremilast

[a]Response rates for deucravacitinib and apremilast were based on treatment efficacy demonstrated in POETYK PSO-1 and PSO-2.
NNT, number needed to treat; PASI 75, 75% reduction from baseline in Psoriasis Area and Severity Index score; sPGA 0/1, static Physician's Global Assessment score of 0 or 1.

Figure 7. Clinical response for deucravacitnib vs 1L branded systemics and NNT for 1L branded systemics

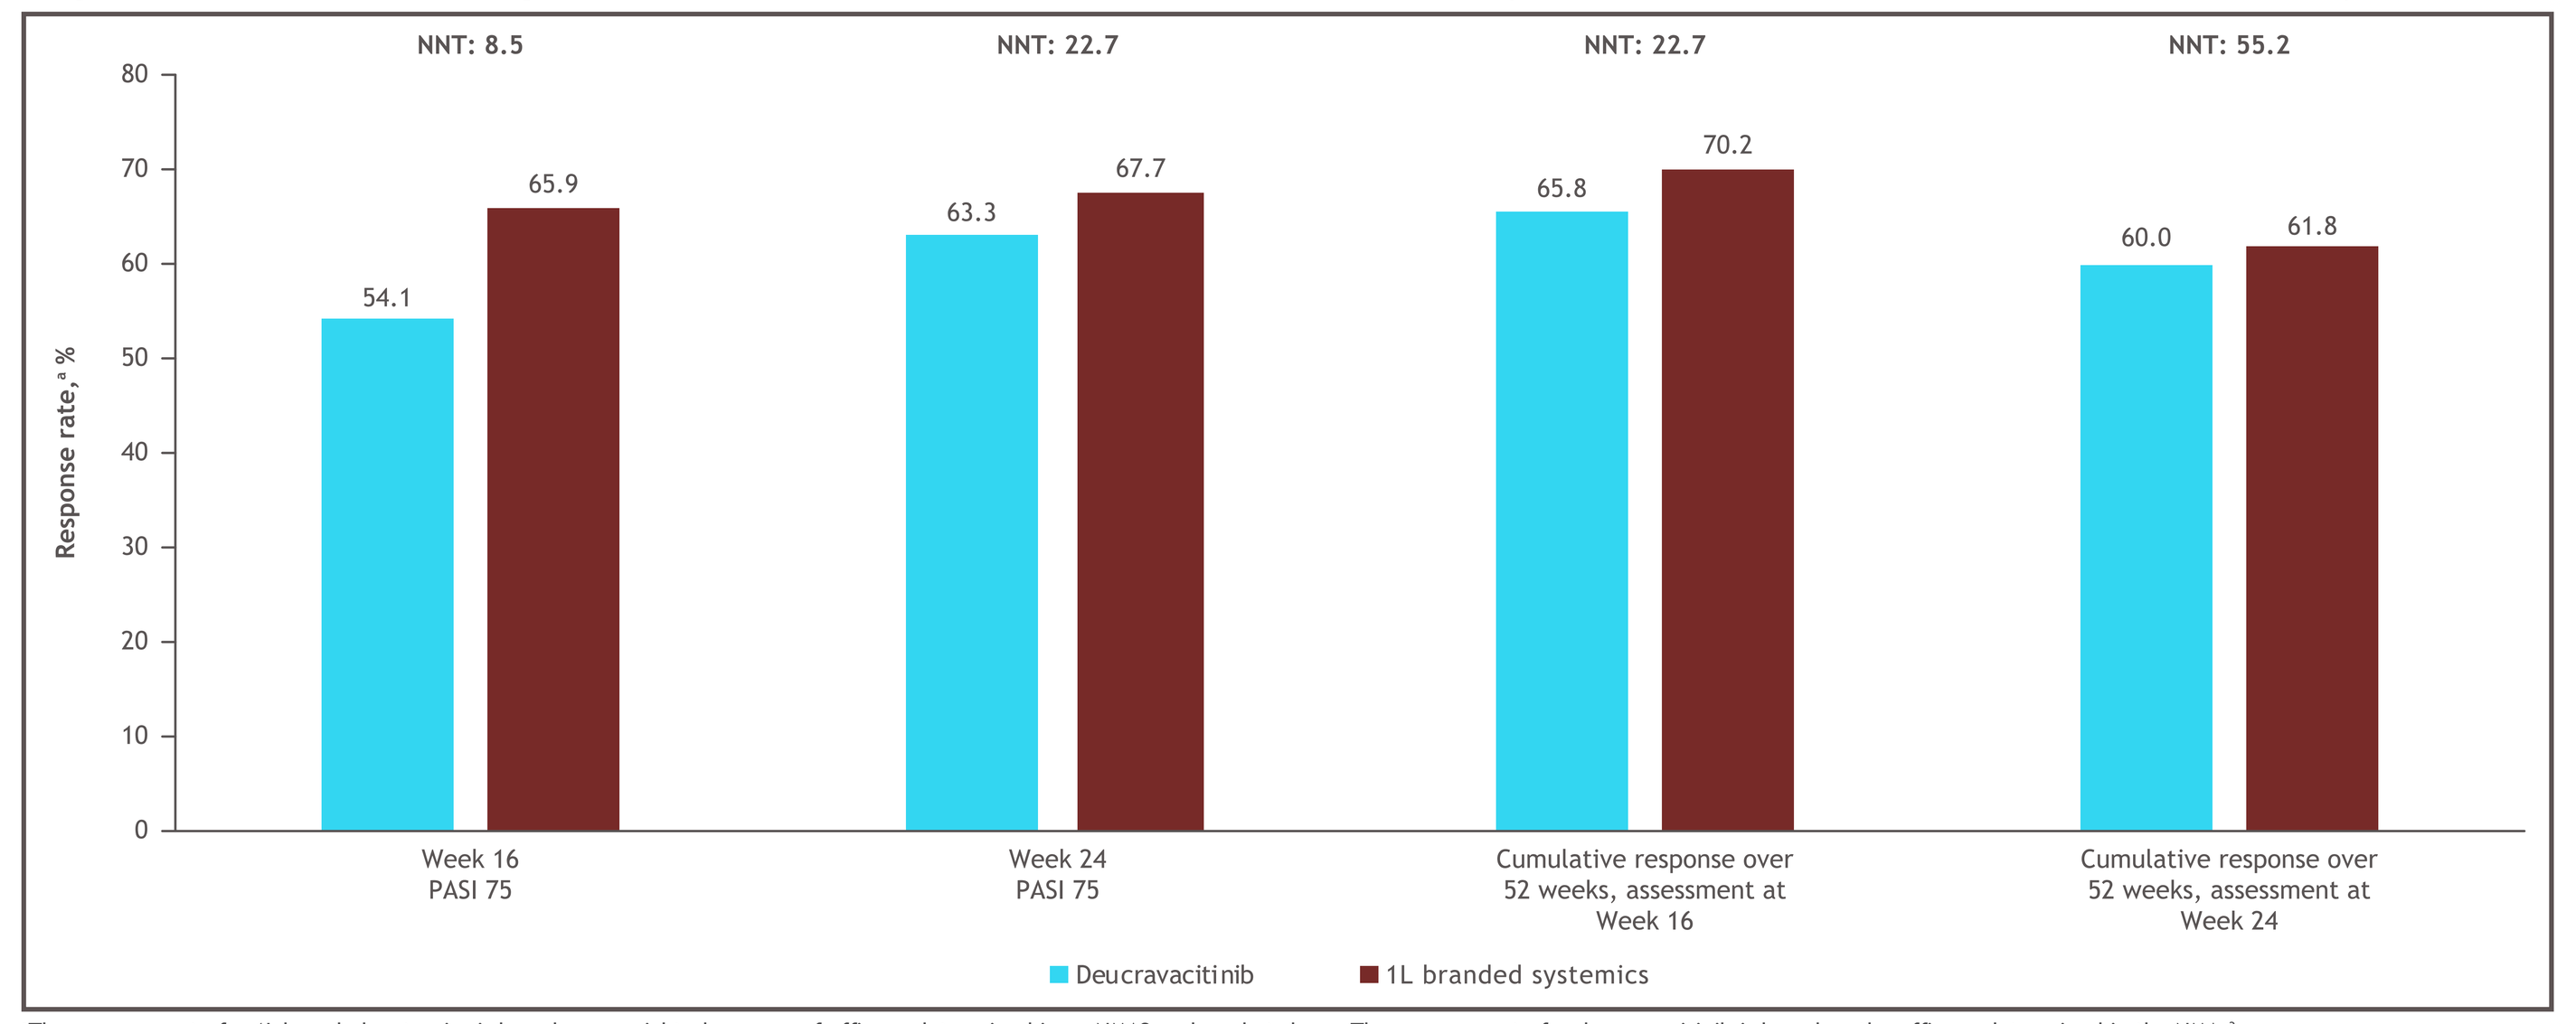

NNT: 8.5
NNT: 22.7
NNT: 22.7
NNT: 55.2
Response rate,[a] %
54.1
65.9
63.3
67.7
65.8
70.2
60.0
61.8
Week 16 PASI 75
Week 24 PASI 75
Cumulative response over 52 weeks, assessment at Week 16
Cumulative response over 52 weeks, assessment at Week 24
Deucravacitinib
1L branded systemics

[a]The response rate for 1L branded systemics is based on a weighted average of efficacy determined in an NMA3 and market share. The response rate for deucravacitinib is based on the efficacy determined in the NMA.[3]
1L, first-line; NNT, number needed to treat; PASI 75, 75% reduction from baseline in Psoriasis Area and Severity Index score; sPGA 0/1, static Physician's Global Assessment score of 0 or 1.

- Scenario analysis confirmed the robustness of results (**Figures 8–11**)

Figure 8. CPR for deucravacitinib vs apremilast: 10% increase in comparator drug prices[a] or no discontinuation rate assumed

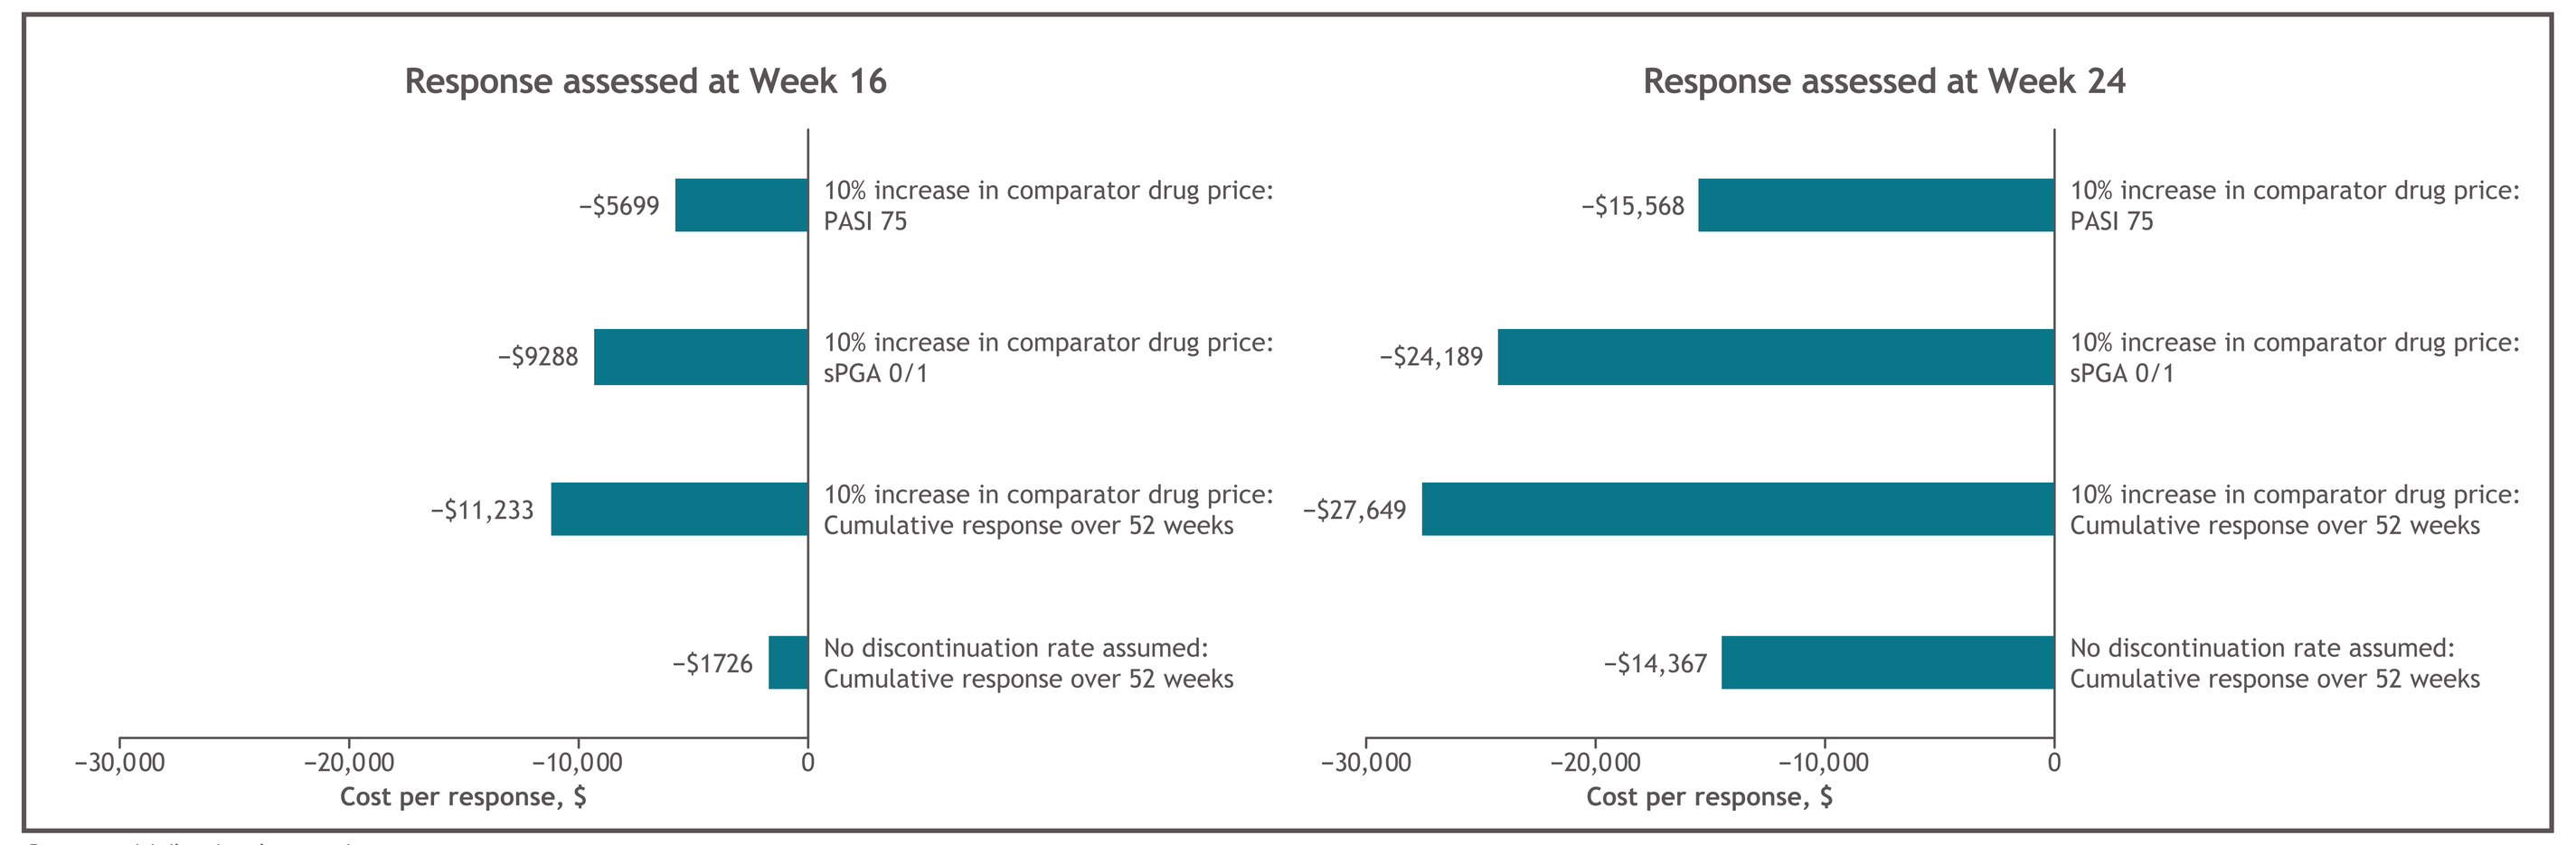

Response assessed at Week 16
−$5699
10% increase in comparator drug price: PASI 75
−$9288
10% increase in comparator drug price: sPGA 0/1
−$11,233
10% increase in comparator drug price: Cumulative response over 52 weeks
−$1726
No discontinuation rate assumed: Cumulative response over 52 weeks
Response assessed at Week 24
−$15,568
−$24,189
−$27,649
−$14,367
Cost per response, $

[a]Deucravacitinib price does not increase.
CPR, cost per response; PASI 75, 75% reduction from baseline in Psoriasis Area and Severity Index score; sPGA 0/1, static Physician's Global Assessment score of 0 or 1.

Figure 9. CPR for deucravacitinib vs apremilast: 2L treatments

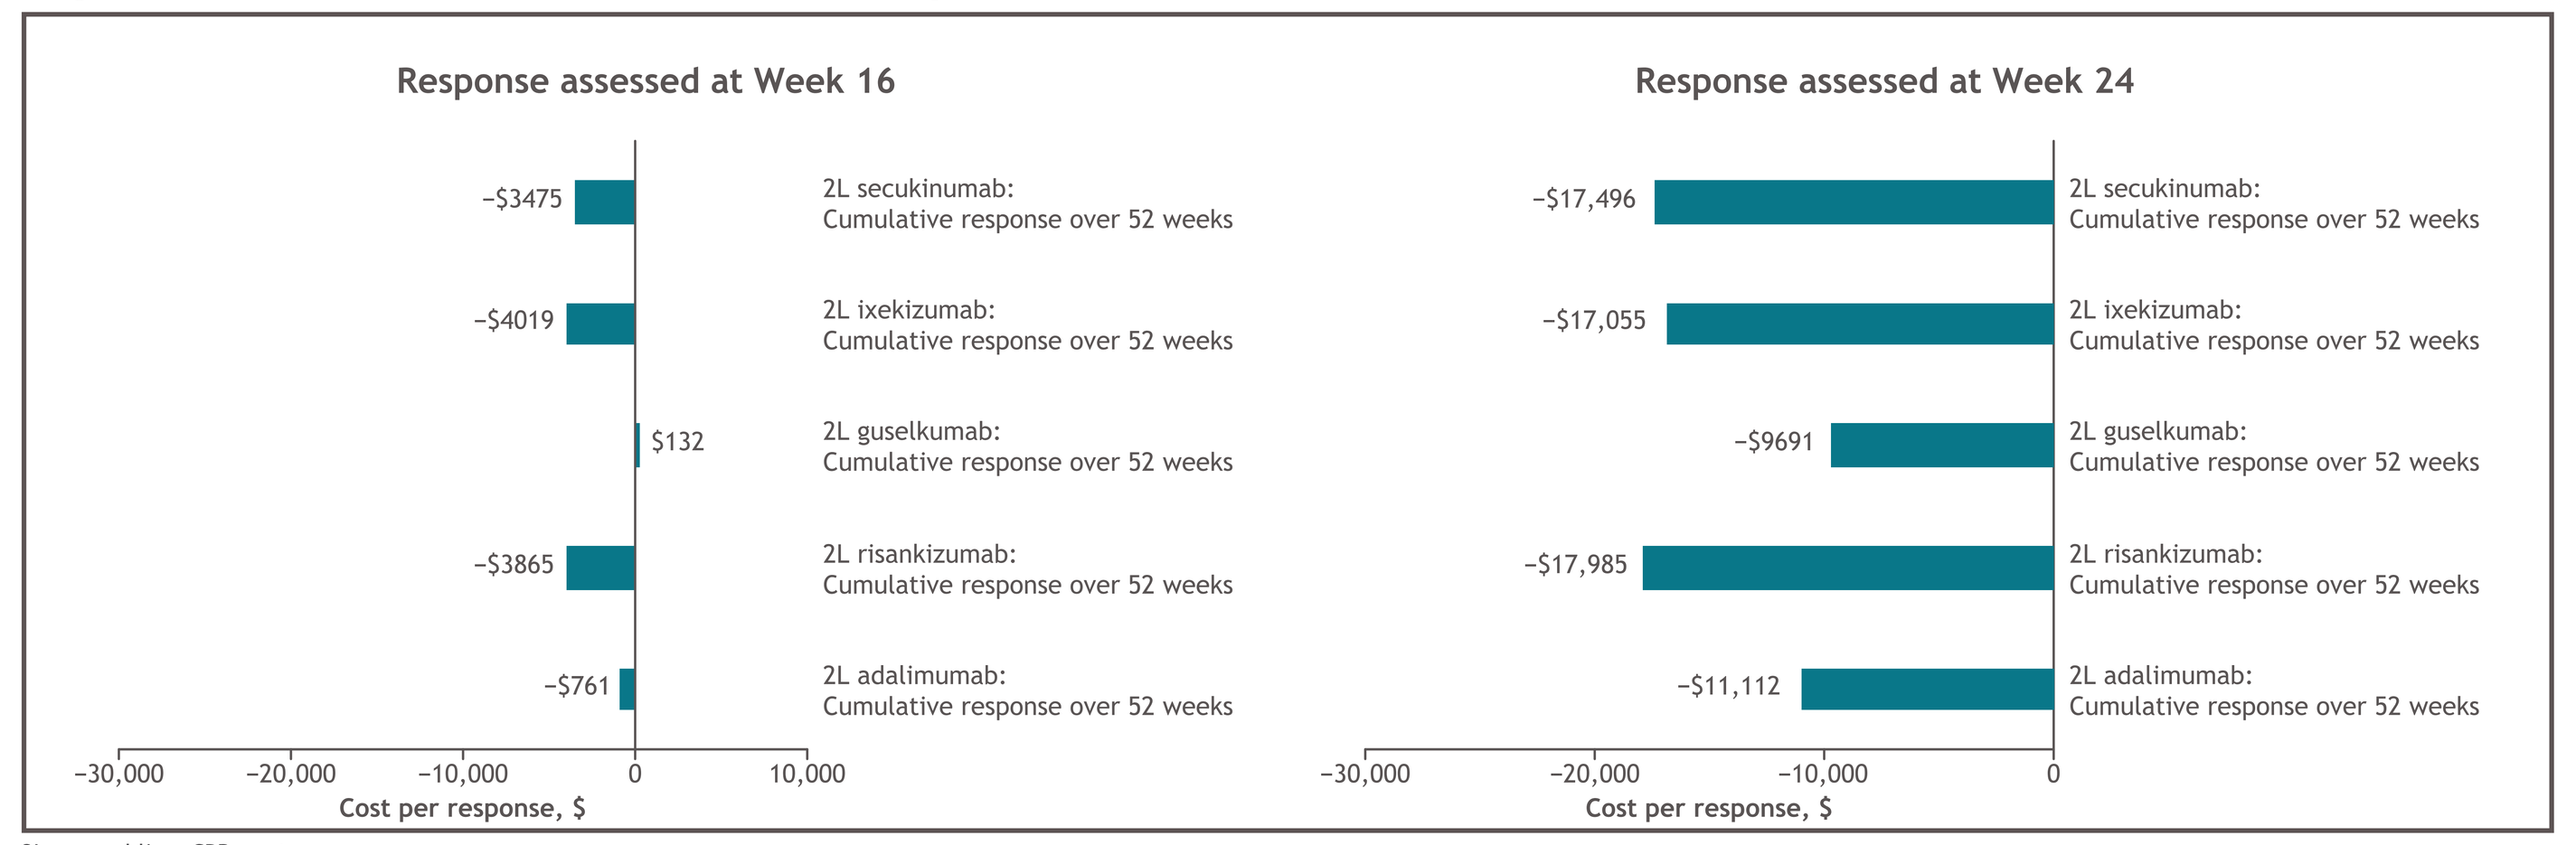

Response assessed at Week 16
−$3475
2L secukinumab: Cumulative response over 52 weeks
−$4019
2L ixekizumab: Cumulative response over 52 weeks
$132
2L guselkumab: Cumulative response over 52 weeks
−$3865
2L risankizumab: Cumulative response over 52 weeks
−$761
2L adalimumab: Cumulative response over 52 weeks
Response assessed at Week 24
−$17,496
−$17,055
−$9691
−$17,985
−$11,112
Cost per response, $

2L, second-line; CPR, cost per response.

Figure 10. CPR for deucravacitinib vs 1L branded systemics: 10% increase in comparator drug prices[a] or no discontinuation rate assumed

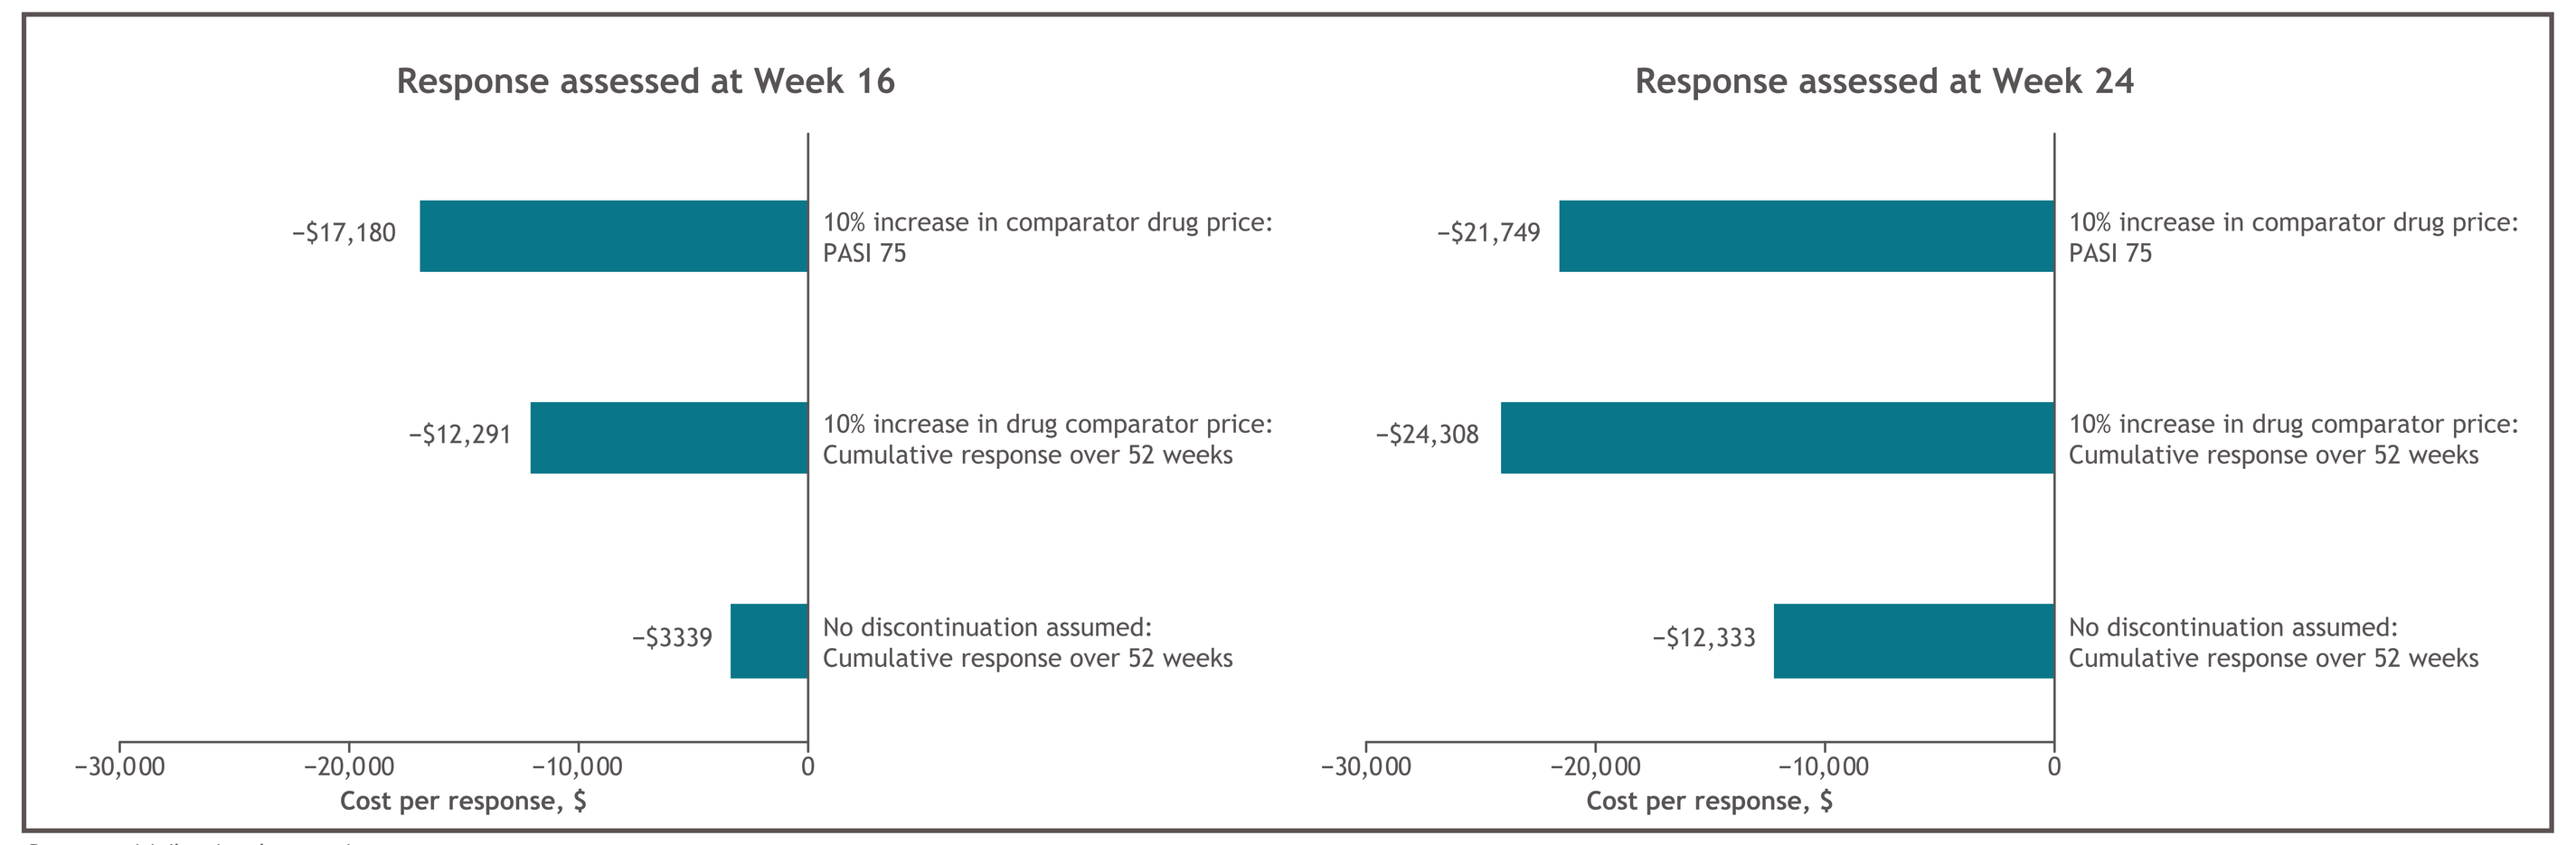

Response assessed at Week 16
−$17,180
10% increase in comparator drug price: PASI 75
−$12,291
10% increase in drug comparator price: Cumulative response over 52 weeks
−$3339
No discontinuation assumed: Cumulative response over 52 weeks
Response assessed at Week 24
−$21,749
−$24,308
−$12,333
Cost per response, $

[a]Deucravacitinib price does not increase.
1L, first line; CPR, cost per response; PASI 75, 75% reduction from baseline in Psoriasis Area and Severity Index score; sPGA 0/1, static Physician's Global Assessment score of 0 or 1.

Figure 11. CPR for deucravacitinib vs 1L branded systemics: 2L treatments

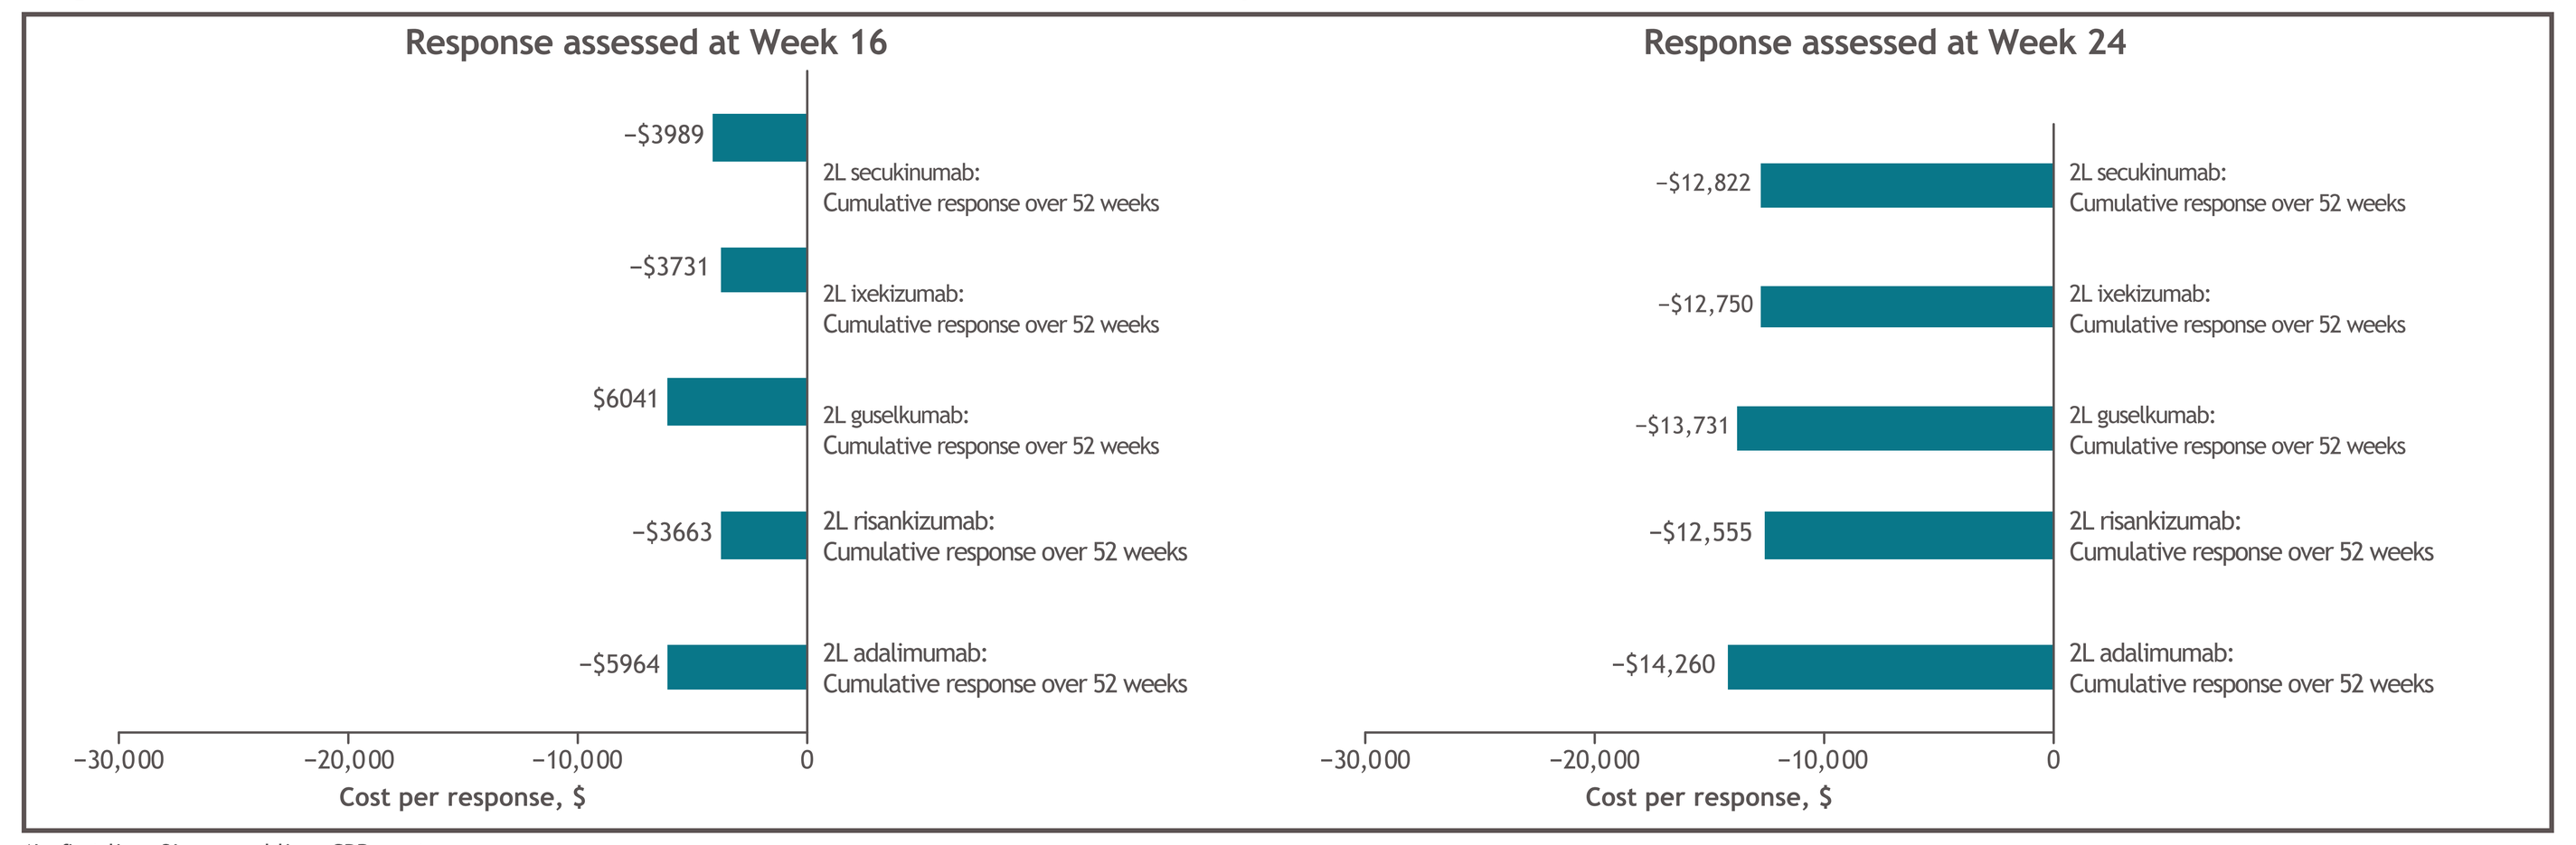

Response assessed at Week 16
−$3989
2L secukinumab: Cumulative response over 52 weeks
−$3731
2L ixekizumab: Cumulative response over 52 weeks
$6041
2L guselkumab: Cumulative response over 52 weeks
−$3663
2L risankizumab: Cumulative response over 52 weeks
−$5964
2L adalimumab: Cumulative response over 52 weeks
Response assessed at Week 24
−$12,822
−$12,750
−$13,731
−$12,555
−$14,260
Cost per response, $

1L, first-line; 2L, second-line; CPR, cost per response.

Conclusion

- For patients with moderate to severe plaque PsO, deucravacitinib is associated with a lower CPR compared with apremilast and 1L branded systemics in both the short term and the long term

References

1. Al Sawah S, et al. *J Med Econ*. 2017;20:982–990. **2.** Sotyktu [package insert]. Princeton, NJ: Bristol Myers Squibb; 2022. **3.** Armstrong AW, et al. [poster a] To be presented at Fall Clinical Dermatology Conference; October 20–23, 2022; Las Vegas, NV. **4.** Blauvelt A, et al. *J Manag Care Spec Pharm*. 2021;27:84–94. **5.** Armstrong AW, et al. *J Dermatol Treat*. 2017;28:492–499. **6.** Armstrong AW, et al. [poster b] To be presented at Fall Clinical Dermatology Conference; October 20–23, 2022; Las Vegas, NV. **7.** Feldman SR, et al. *Dermatol Online J*. 2021;27:13030/qt03t0s9j6.

Acknowledgments

- This study was sponsored by Bristol Myers Squibb
- Laetitia N'Dri of Bristol Myers Squibb, Princeton, NJ, assisted the authors with the preparation of the data presented
- Writing and editorial assistance was provided by Tulika Bhushan Bahukhandi, RPh, MS, of Peloton Advantage, LLC, an OPEN Health company, and funded by Bristol Myers Squibb

Disclosures

- **SP, SH, KW,** and **VP:** Employees of and may own stock options in Bristol Myers Squibb
- **ML, JS,** and **DP:** Employees of OPEN Health, which received consulting fees from Bristol Myers Squibb

Presented at the Dermatology Innovation Symposium; October 14–16, 2022; Anaheim, CA.

Email for Sang Hee Park, MPH: Sarah.Park@bms.com

This poster may not be reproduced without written permission from the authors.